Real-World Biologic Treatment Prescribing Patterns and Reasons for Discontinuation in Patients With UC and CD

Abbey Nakano, PharmD[1]; *Dave Iwanyckyj, BA[1]; Fernando Otalora[1]; John Perez[1]; Melanie Jardim, PhD[1]
[1]Amplity, Inc, Langhorne, PA, USA

amplity

RWD103

Background

- Inflammatory bowel disease (IBD), which includes ulcerative colitis (UC) and Crohn's disease (CD), affects an estimated 2.4 million Americans[1]
- Although biologic therapies for patients with UC and CD have improved outcomes in patients with moderate-to-severe IBD, patients often lose response to therapy or may experience side effects, and ultimately require additional treatment options[2]
- Selection of first-line biologic therapy for patients with moderate-to-severe IBD, and the decision to switch to an alternative biologic, are typically based on an individualized approach and shared decision-making between the physician and patient[3]
- This study aimed to assess real-world prescribing patterns of biologics and explore factors driving therapy change

Methods

- This retrospective observational study used natural language processing (NLP) to search and analyze the Amplity AnswerY™ database and platform for patients diagnosed with UC or CD
- Amplity AnswerY™ is Amplity's real-world database and platform built from HIPAA-compliant transcriptions of US prescriber–patient visits. Using AI and NLP, it extracts, visualizes, and summarizes treatment discussions and clinical decisions. Covering inpatient and outpatient care across 70+ specialties since 2017, AnswerY was known as Amplity Insights™ prior to January 2025
- AnswerY was queried for patients with a diagnosis of UC or CD who were prescribed at least 1 biologic therapy
- Patients were followed through subsequent lines of biologic therapy until documented discontinuation of therapy or no further mention of a subsequent biologic therapy
- Trends in prescribing habits were gathered for all-time records, then split by records before 2023 and 2023 onward to understand influence of biosimilar adoption on prescribing habits
- Reasons for discontinuation were documented between lines of therapy

Conclusion

- Analysis of the AnswerY database shows adalimumab and infliximab are the most utilized first-line biologics, with a wider range of agents used in second-line therapy
- Using AI and NLP to review patient–provider transcripts, AnswerY was able to track prescribing trends through lines of therapy and identify reasons for treatment discontinuation
- The most common reasons for discontinuation of first-line biologic therapy were adverse events and efficacy
- This real-world data can be used to further understand the treatment landscape of biologics in the treatment of IBD

*PRESENTING AUTHOR

DISCLOSURES
All authors are employees of Amplity, Inc.

To learn more about this study and further research by Amplity AnswerY™, please scan this code.

REFERENCES
1. Lewis JD, et al. Incidence, prevalence, and racial and ethnic distribution of inflammatory bowel disease in the United States. *Gastroenterology.* 2023;165:1197-1205.e2.
2. Kapizioni C, et al. Biologic therapy for inflammatory bowel disease: real-world comparative effectiveness and impact of drug sequencing in 13 222 patients within the UK IBD BioResource. *J Crohns Colitis.* 2024;18:790-800.
3. Song K, Wu D. Shared decision-making in the management of patients with inflammatory bowel disease. *World J Gastroenterol.* 2022;28:3092-3100.

Results: Demographics for Patients With UC and CD

- AnswerY identified 8738 patients with UC (**Figure 1**) and 20,223 patients with CD (**Figure 2**) treated with biologic therapy
- Overall, as seen in **Figures 3** and **4**, there was a wide range of age groups, slightly more female patients (51.93% UC, 56.64% CD), and a majority of patients were Caucasian (89.28% UC, 88.09% CD)
- The geographic distribution of patient records is shown in **Figure 5** and **Figure 6**

Figure 1: Number of Patients and Records With Mention of UC and Biologic Therapy

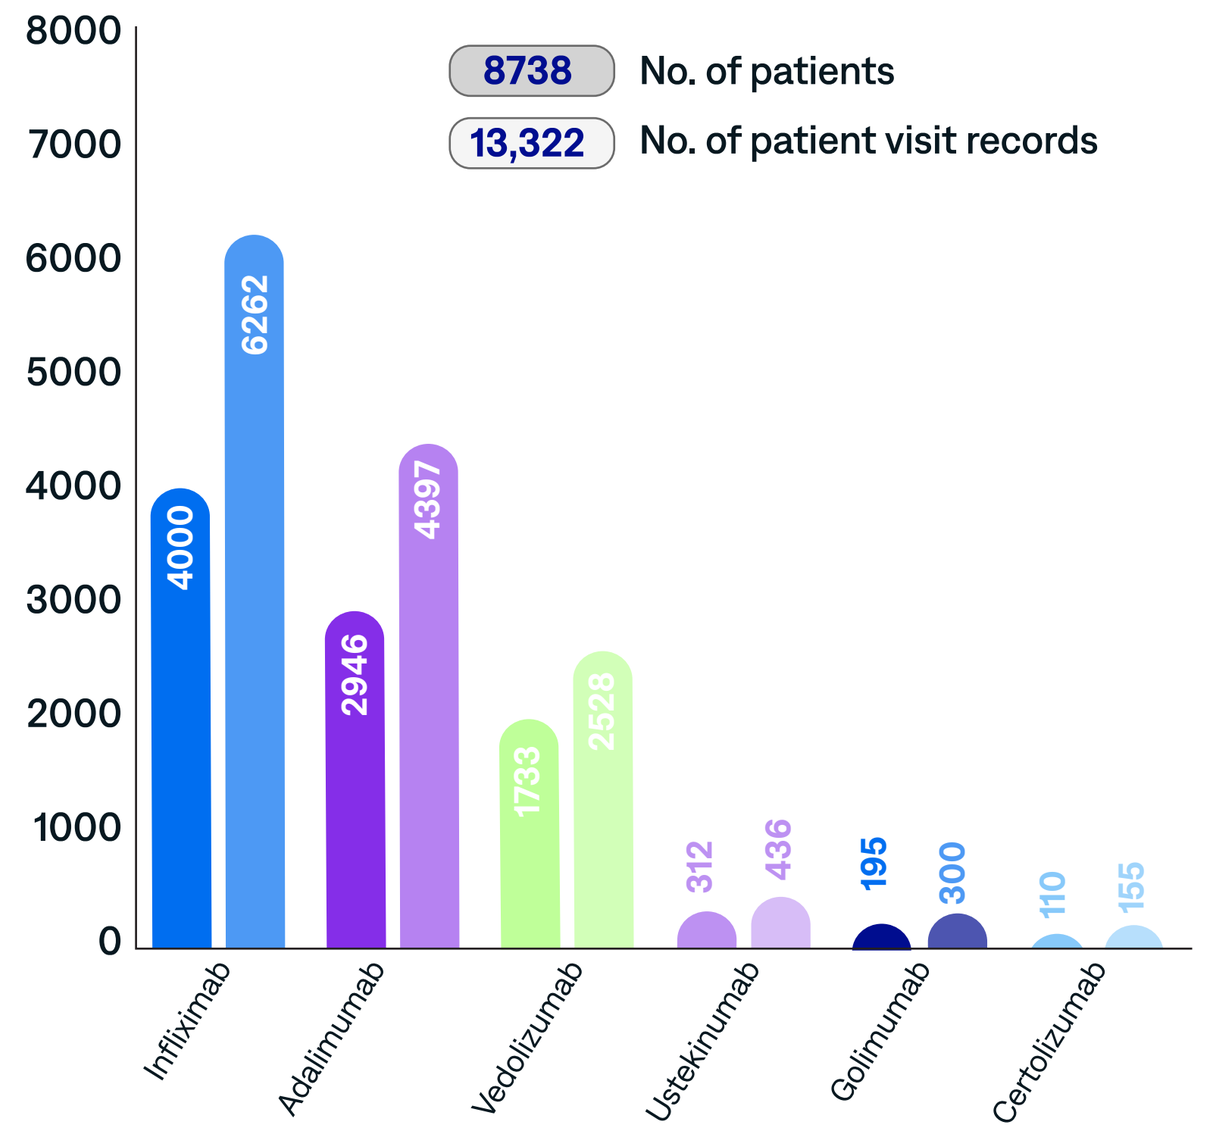
8738 No. of patients
13,322 No. of patient visit records
8000
7000
6000
5000
4000
3000
2000
1000
0
4000
6262
2946
4397
1733
2528
312
436
195
300
110
155
Infliximab
Adalimumab
Vedolizumab
Ustekinumab
Golimumab
Certolizumab

UC, ulcerative colitis.

Figure 2: Number of Patients and Records With Mention of CD and Biologic Therapy

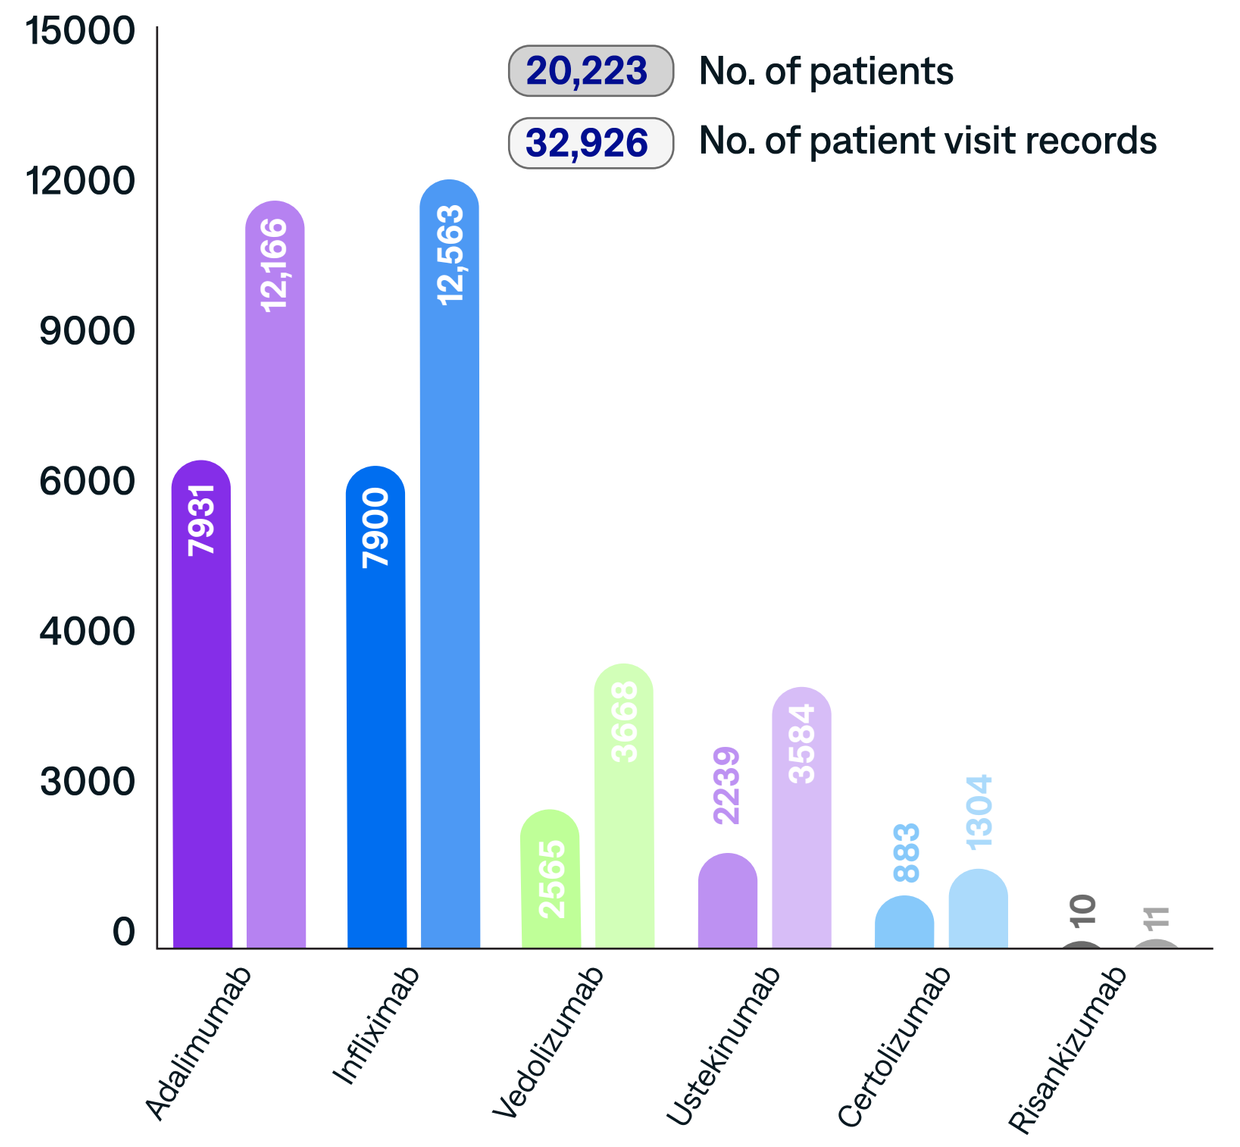
20,223 No. of patients
32,926 No. of patient visit records
15000
12000
9000
6000
4000
3000
0
7931
12,166
7900
12,563
2565
3668
2239
3584
883
1304
10
11
Adalimumab
Infliximab
Vedolizumab
Ustekinumab
Certolizumab
Risankizumab

CD, Crohn's disease.

Figure 3: Demographics for Patients With UC

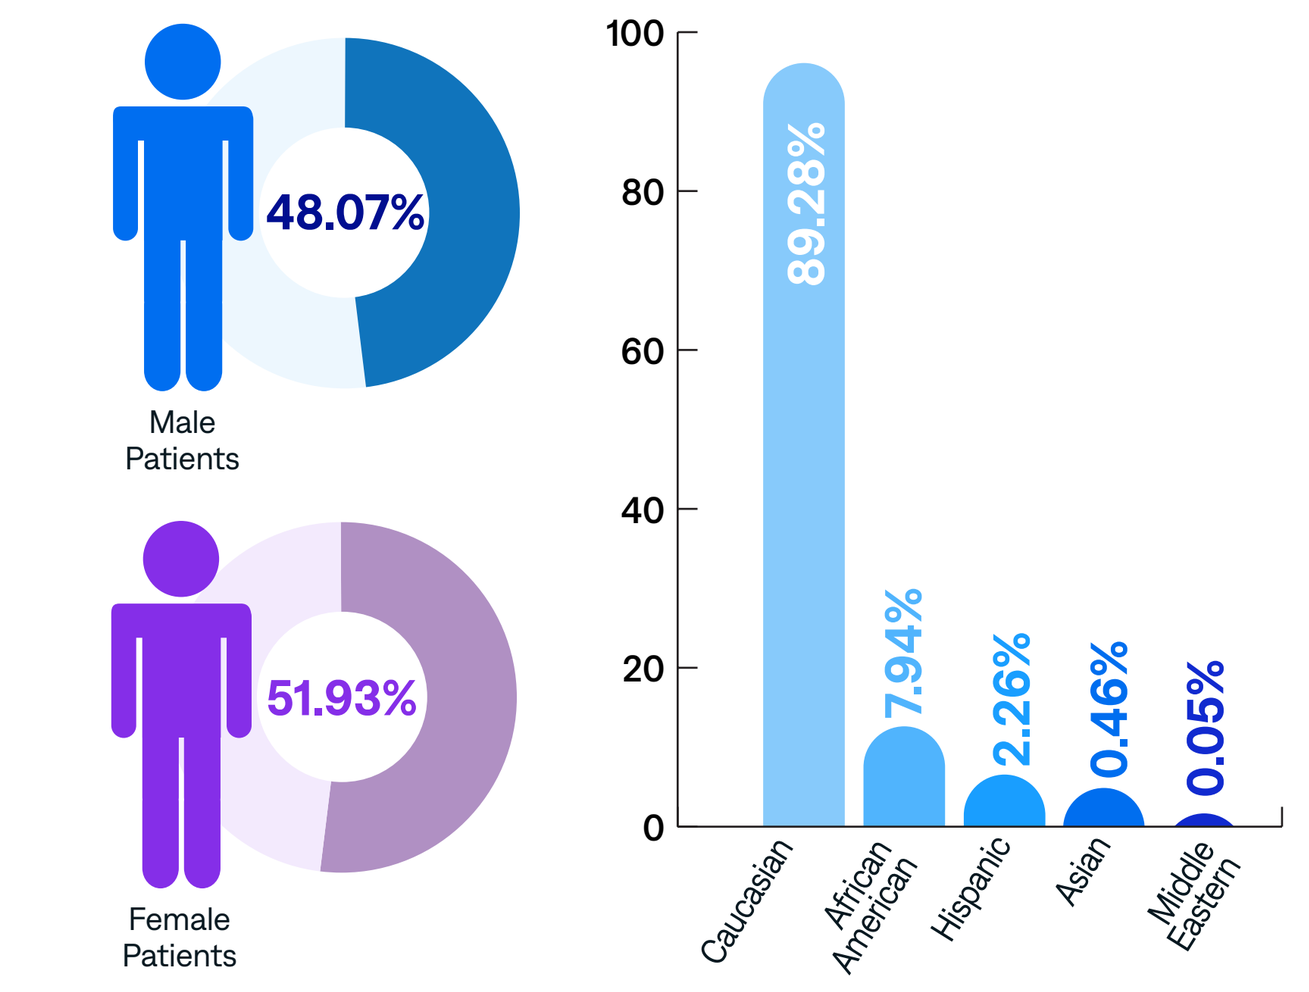
48.07%
Male Patients
51.93%
Female Patients
100
80
60
40
20
0
89.28%
7.94%
2.26%
0.46%
0.05%
Caucasian
African American
Hispanic
Asian
Middle Eastern

UC, ulcerative colitis.

Figure 4: Demographics for Patients With CD

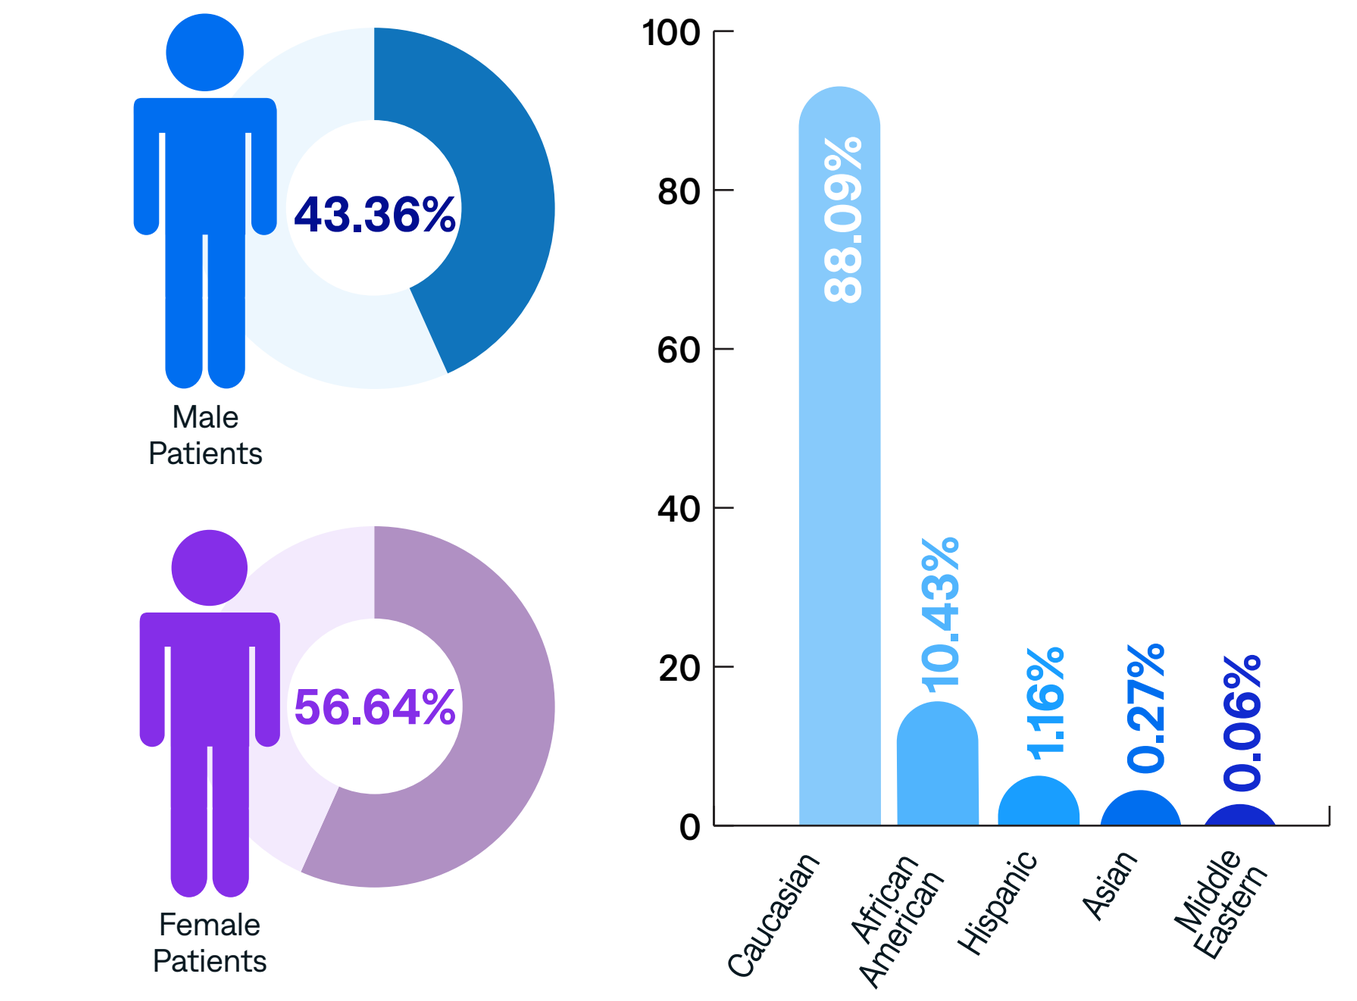
43.36%
Male Patients
56.64%
Female Patients
100
80
60
40
20
0
88.09%
10.43%
1.16%
0.27%
0.06%
Caucasian
African American
Hispanic
Asian
Middle Eastern

CD, Crohn's disease.

Figure 5: Geographic Distribution of Patients With UC

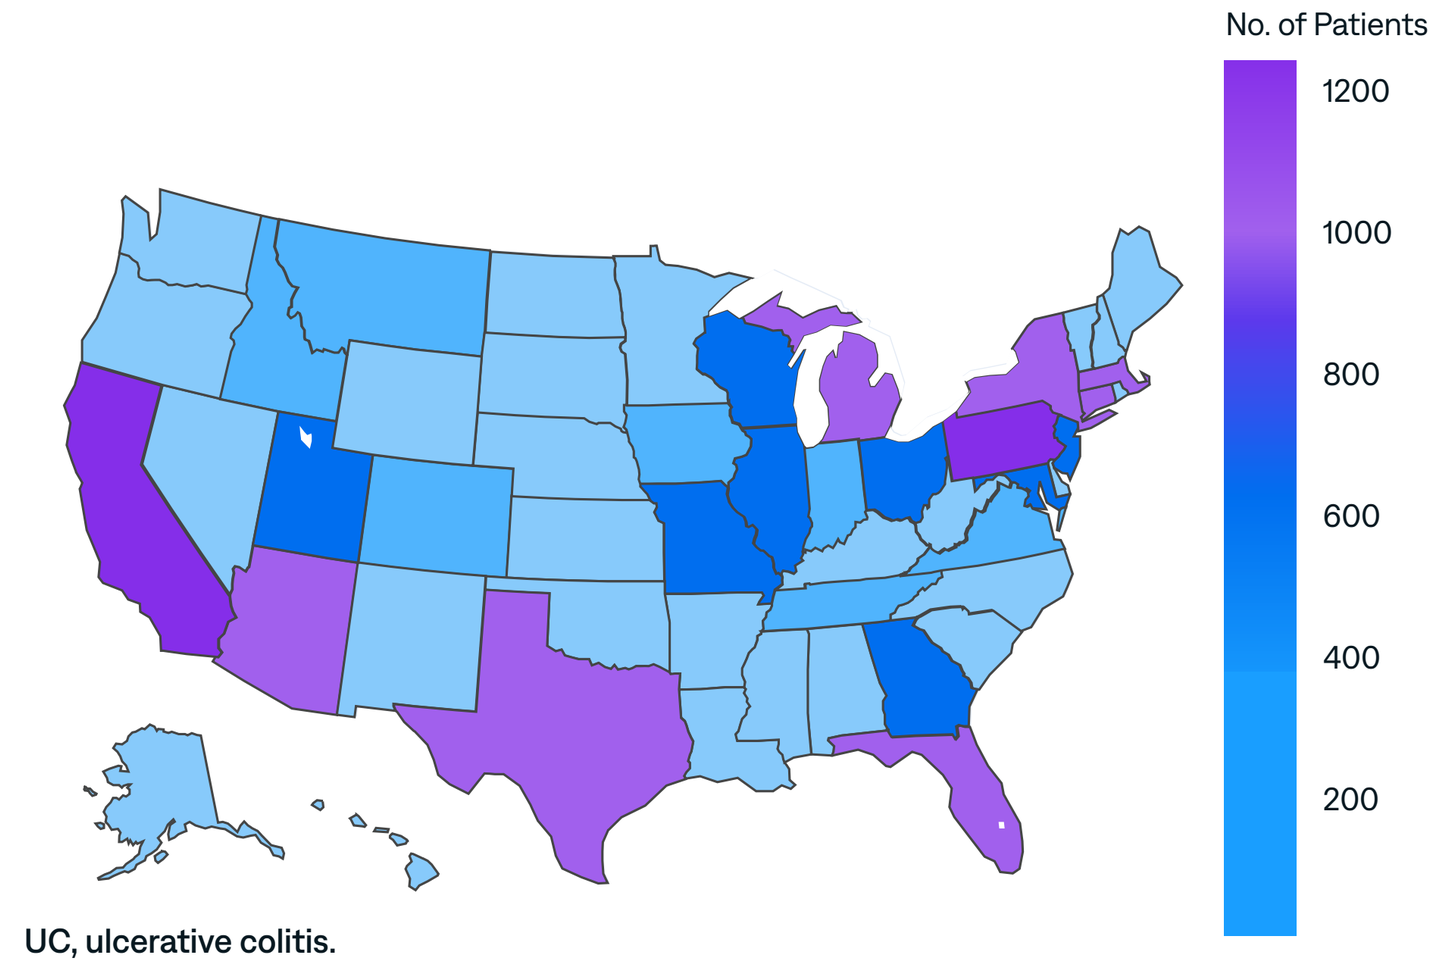
No. of Patients
1200
1000
800
600
400
200

UC, ulcerative colitis.

Figure 6: Geographic Distribution of Patients With CD

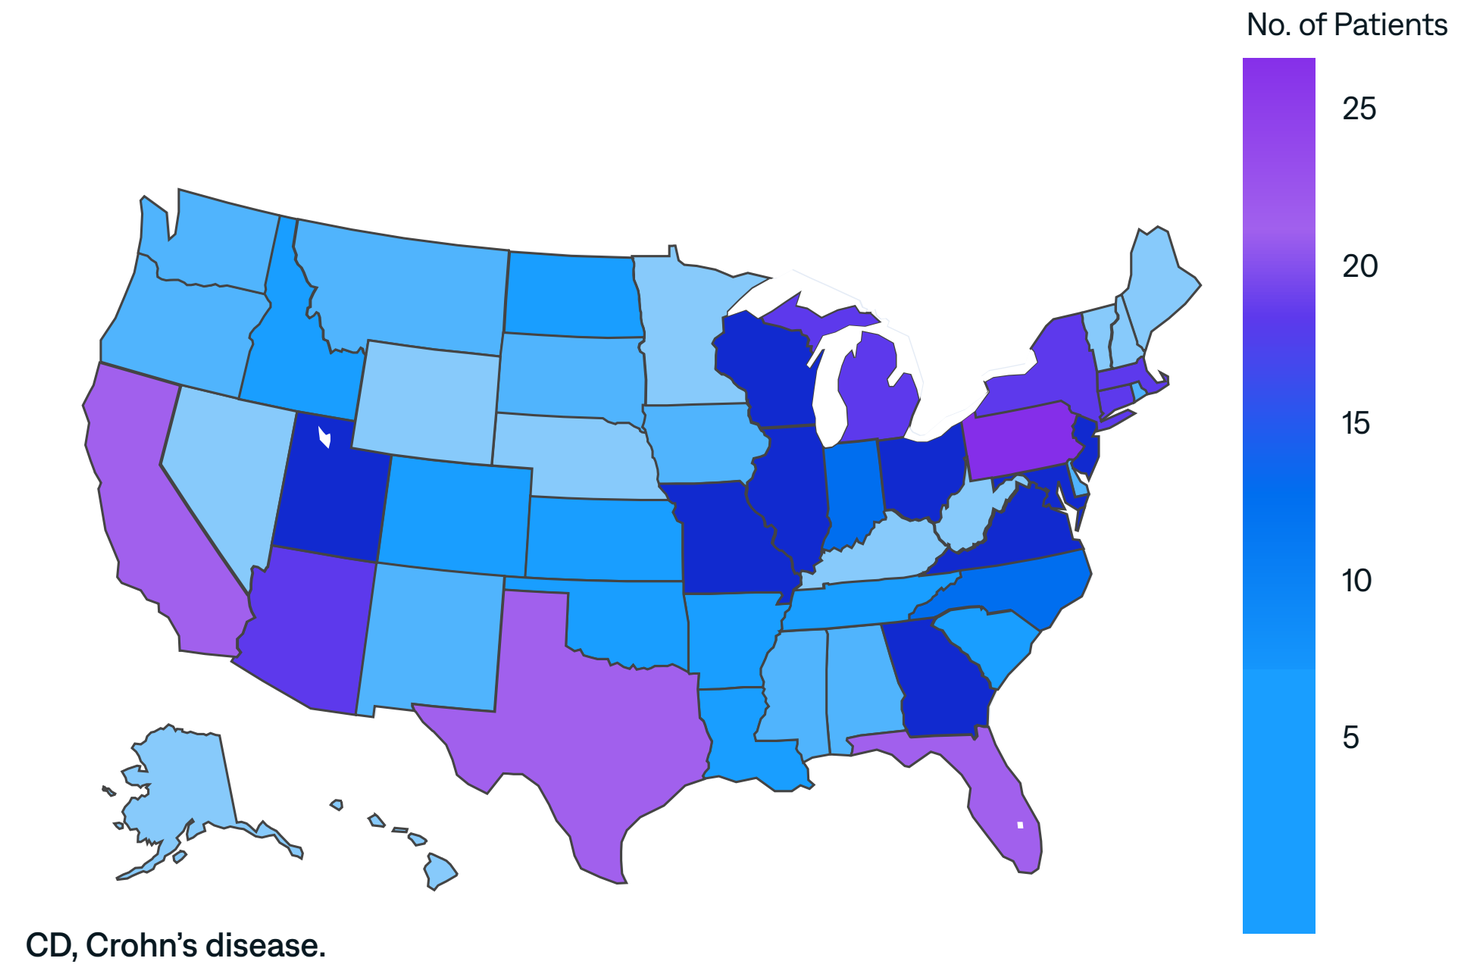
No. of Patients
25
20
15
10
5

CD, Crohn's disease.

Among patients with UC or CD, the most commonly prescribed first-line biologics were adalimumab and infliximab, with more variability in the second line

Results: Biologic Treatment Journey for Patients With UC

- All-time, the most common first-line biologics prescribed for patients with UC were adalimumab (41.21%) and infliximab (36.00%), followed by vedolizumab (15.60%) (**Figure 7**)
- Second-line therapy for patients with UC included infliximab (33.30%), vedolizumab (30.40%), and adalimumab (23.50%) (**Figure 7**)
- These trends were similar for ≤2022 and ≥2023

Figure 7: Biologic Treatment Sequencing for Patients With UC

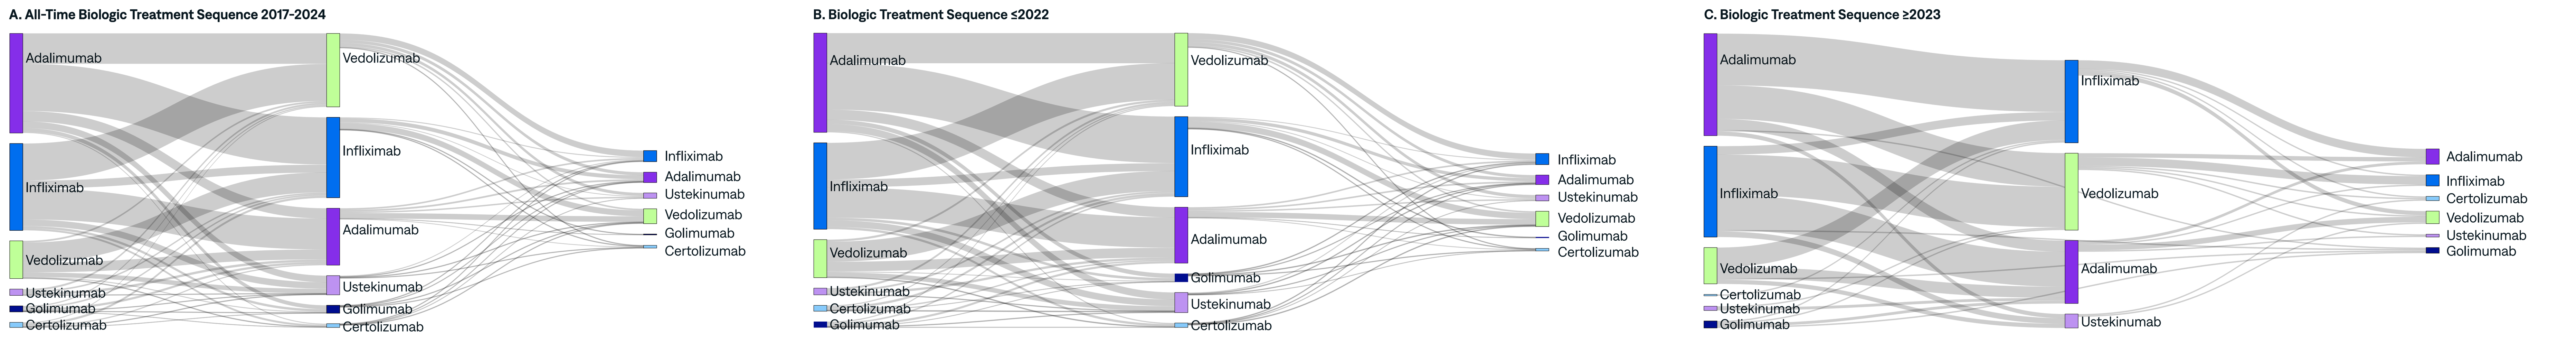
A. All-Time Biologic Treatment Sequence 2017-2024
Adalimumab
Infliximab
Vedolizumab
Ustekinumab
Golimumab
Certolizumab
Vedolizumab
Infliximab
Adalimumab
Ustekinumab
Golimumab
Certolizumab
Infliximab
Adalimumab
Ustekinumab
Vedolizumab
Golimumab
Certolizumab
B. Biologic Treatment Sequence ≤2022
Adalimumab
Infliximab
Vedolizumab
Ustekinumab
Certolizumab
Golimumab
Vedolizumab
Infliximab
Adalimumab
Golimumab
Ustekinumab
Certolizumab
Infliximab
Adalimumab
Ustekinumab
Vedolizumab
Golimumab
Certolizumab
C. Biologic Treatment Sequence ≥2023
Adalimumab
Infliximab
Vedolizumab
Certolizumab
Ustekinumab
Golimumab
Infliximab
Vedolizumab
Adalimumab
Ustekinumab
Adalimumab
Infliximab
Certolizumab
Vedolizumab
Ustekinumab
Golimumab

UC, ulcerative colitis.

Results: Biologic Treatment Journey for Patients With CD

- All-time, the most common first-line biologics in patients with CD were adalimumab (39.70%) and infliximab (32.60%), followed by vedolizumab (32.20%) (**Figure 8**)
- Second-line therapy was distributed among infliximab (28.50%), adalimumab (26.00%), ustekinumab (19.80%), and vedolizumab (19.67%) (**Figure 8**)
- These trends were similar for ≤2022 and ≥2023

Figure 8: Biologic Treatment Sequencing for Patients With CD

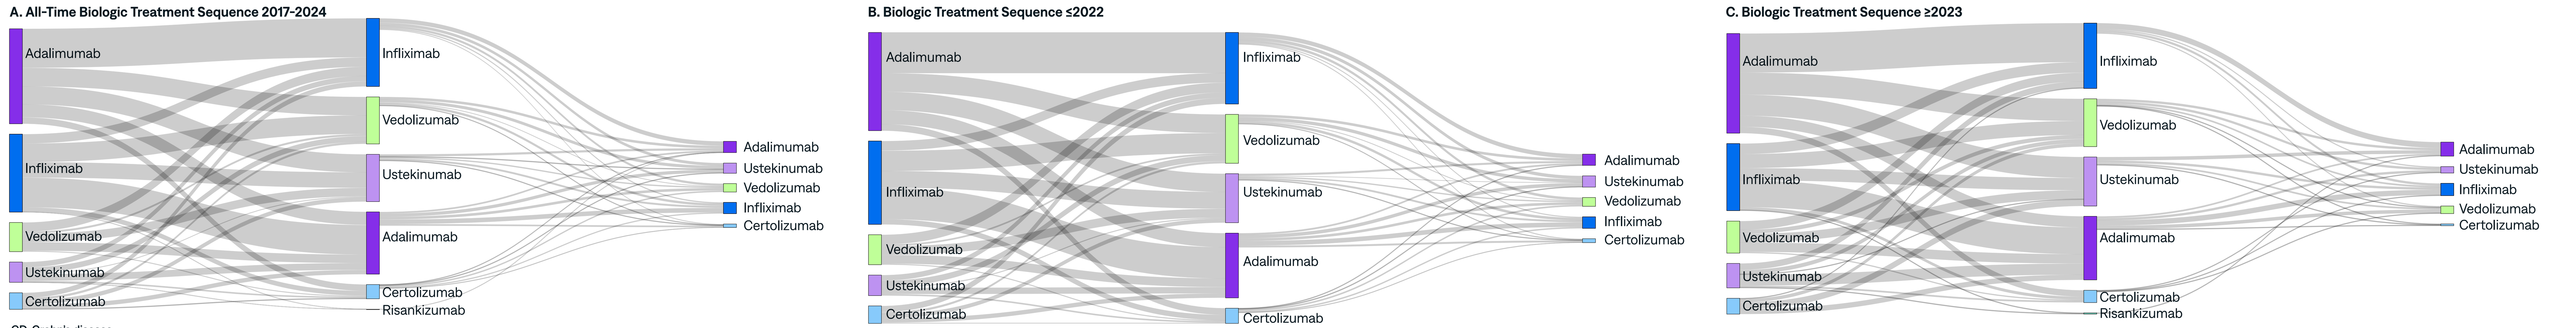
A. All-Time Biologic Treatment Sequence 2017-2024
Adalimumab
Infliximab
Vedolizumab
Ustekinumab
Certolizumab
Infliximab
Vedolizumab
Ustekinumab
Adalimumab
Certolizumab
Risankizumab
Adalimumab
Ustekinumab
Vedolizumab
Infliximab
Certolizumab
B. Biologic Treatment Sequence ≤2022
Adalimumab
Infliximab
Vedolizumab
Ustekinumab
Certolizumab
Infliximab
Vedolizumab
Ustekinumab
Adalimumab
Certolizumab
Adalimumab
Ustekinumab
Vedolizumab
Infliximab
Certolizumab
C. Biologic Treatment Sequence ≥2023
Adalimumab
Infliximab
Vedolizumab
Ustekinumab
Certolizumab
Infliximab
Vedolizumab
Ustekinumab
Adalimumab
Certolizumab
Risankizumab
Adalimumab
Ustekinumab
Infliximab
Vedolizumab
Certolizumab

CD, Crohn's disease.

Adverse events and efficacy were the most common reasons for treatment discontinuation

Results: Treatment Discontinuation

- The most frequently reported reasons for discontinuing first-line biologic therapy were adverse events and efficacy (**Table 1** and **Table 2**)
- In patients with UC, first-line vedolizumab was discontinued due to adverse events more often than first-line infliximab and adalimumab
- In patients with CD, vedolizumab was discontinued due to patient preference more often than first-line infliximab and adalimumab

Table 1: Reasons for Discontinuation From First- to Second-Line Therapy in Patients With UC

1L → 2L	Adverse events (%)	Efficacy (%)	Insurance (%)	Patient preference (%)	Cost (%)
Adalimumab → Infliximab (n=98)	20.08	20.08	n/a	n/a	0.84
Adalimumab → Vedolizumab (n=67)	17.53	19.48	1.30	5.19	n/a
Infliximab → Adalimumab (n=77)	28.66	15.29	0.64	3.82	0.64
Infliximab → Vedolizumab (n=91)	25.00	20.74	n/a	1.06	1.60
Vedolizumab → Adalimumab (n=26)	34.00	14.00	n/a	2.00	2.00
Vedolizumab → Infliximab (n=56)	35.64	16.83	0.99	1.98	n/a

1L, first line; 2L, second line; n/a, not applicable; UC, ulcerative colitis.

Table 2: Discontinuation From First- to Second-Line Therapy in Patients With CD

1L → 2L	Adverse events (%)	Efficacy (%)	Insurance (%)	Patient preference (%)	Cost (%)
Adalimumab → Infliximab (n=161)	19.31	12.23	0.43	1.50	1.07
Adalimumab → Vedolizumab (n=102)	21.27	21.72	2.71	0.45	n/a
Infliximab → Adalimumab (n=161)	28.81	11.58	0.85	2.54	1.69
Infliximab → Vedolizumab (n=106)	24.66	21.97	n/a	0.90	n/a
Vedolizumab → Adalimumab (n=49)	29.70	11.88	n/a	6.93	n/a
Vedolizumab → Infliximab (n=62)	32.73	15.45	1.82	5.45	0.91

1L, first line; 2L, second line; CD, Crohn's disease; n/a, not applicable.